Abstract #114
PB-065

Anti-tumor activity of tarloxotinib, a hypoxia-activated EGFR/HER2 TKI, in HER2 driven cell lines

Adriana Estrada-Bernal, Anh T. Le, Andrea Doak, and Robert C. Doebele
Department of Medicine, Division of Medical Oncology , University of Colorado - Anschutz Medical Campus, Aurora, CO;

School of Medicine
UNIVERSITY OF COLORADO
ANSCHUTZ MEDICAL CAMPUS

Introduction

- NSCLC is the leading cause of cancer-related deaths,[1] but strategies using oncogene-targeted therapies have led to improvements in clinical outcomes for subsets of patients.
- Aberrant *HER2* expression (gene amplifications, mutations or overexpression) in NSCLC and other cancers can cause constitutive activation and downstream signaling.[2] *HER2* gene amplification is present in 2-4% of NSCLC, while *HER2* activating mutations are present in 3% of NSCLC.[3]
- Other oncogenic alterations such as *NRG1* gene fusions and EGFR kinase domain duplications may be amenable to HER2/EGFR kinase inhibition.[4,5]
- *HER2* alterations in lung cancer showed resistance to known TKIs, thus, new therapies need to be developed for lung cancer patients with *HER2* aberrations.[6]
- Solid tumors, including lung cancer, harbor regions of hypoxia[7] which can lead to resistance to radiotherapy, chemotherapy, and immunotherapy. To transform this poor risk feature into a vulnerability, tarloxotinib was designed as a prodrug that is activated under hypoxic conditions to release an irreversible EGFR/HER2 inhibitor (tarloxotinib-E).[8]
- Tarloxotinib can increase the therapeutic ratio over conventional anti-EGFR/HER2 therapies by inhibiting EGFR and HER2 with greater dose-intensity in hypoxic tumors.
- In this study, we evaluate the effect of tarloxotinib on cell lines harboring HER2 alterations, EGFR kinase domain duplication and a *DOC4-NRG1* fusion.

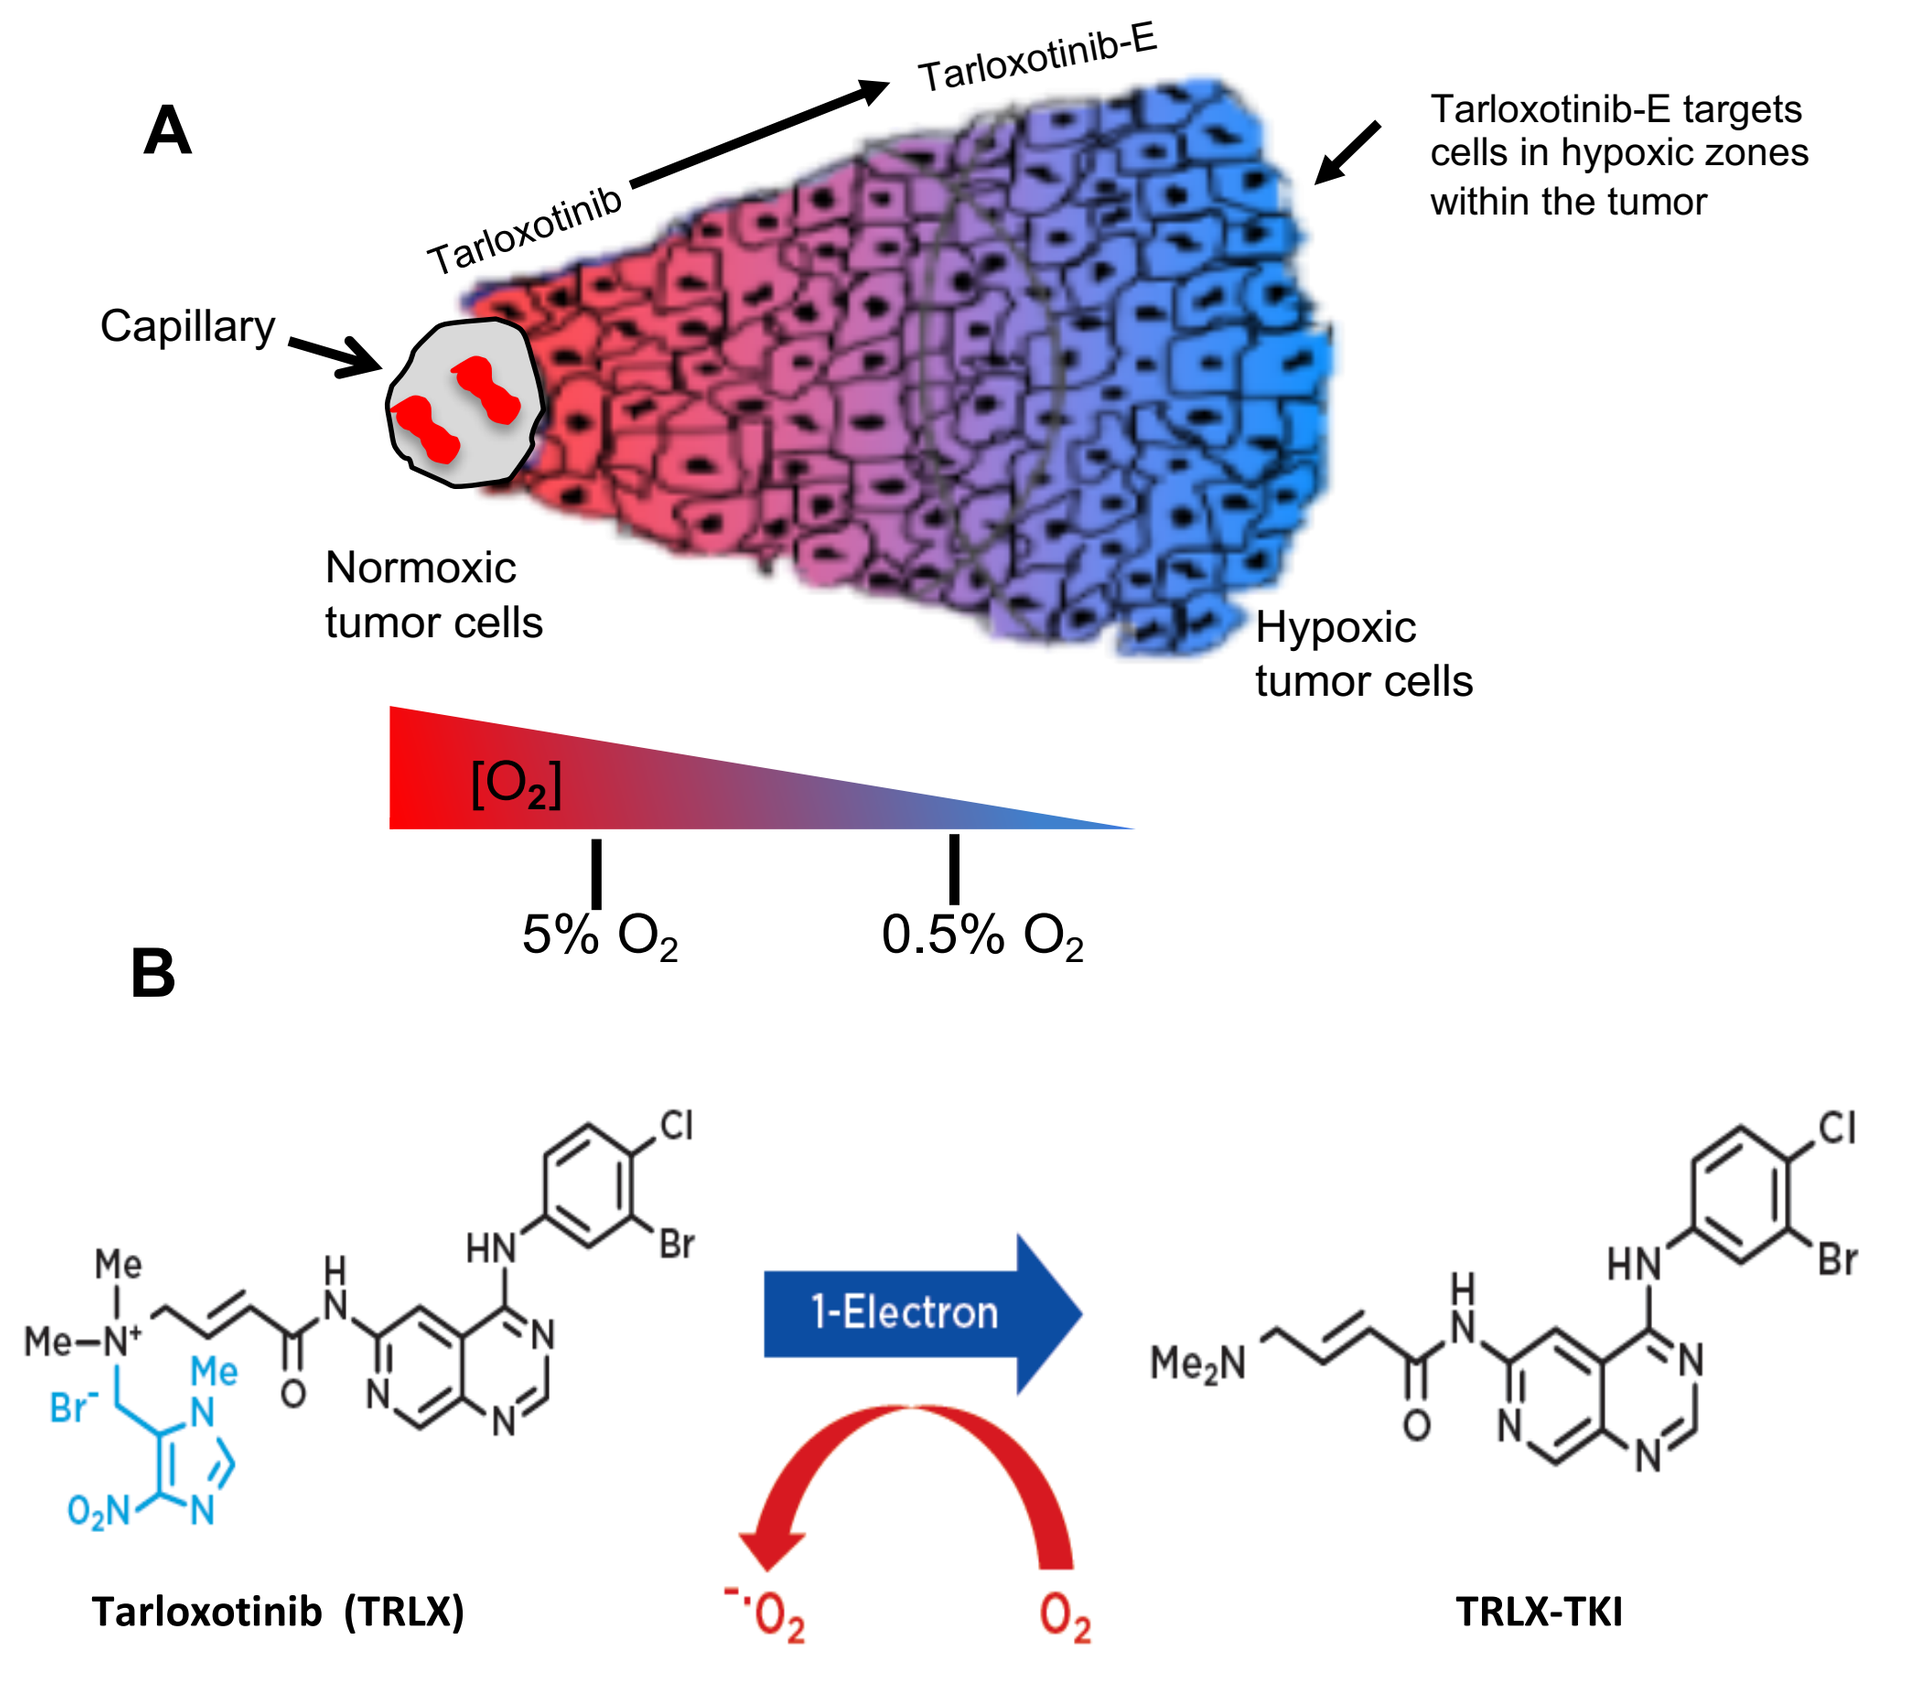
A
Tarloxotinib-E
Tarloxotinib
Tarloxotinib-E targets cells in hypoxic zones within the tumor
Capillary
Normoxic tumor cells
Hypoxic tumor cells
[O2]
5% O2
0.5% O2
B
1-Electron
Tarloxotinib (TRLX)
TRLX-TKI

Figure 1. A) Schematic of hypoxia regions in tumor tissue indicating the areas where tarloxotinib is converted to its active form. **B)** Representation of tarloxotinib conversion to its irreversible EGFR/HER2 inhibitor. Addition of a hypoxia trigger (blue) to tarloxotinib significantly reduces the potency of the prodrug, allowing for administration at higher relative concentrations than the cognate TKI (tarloxotinib-E).

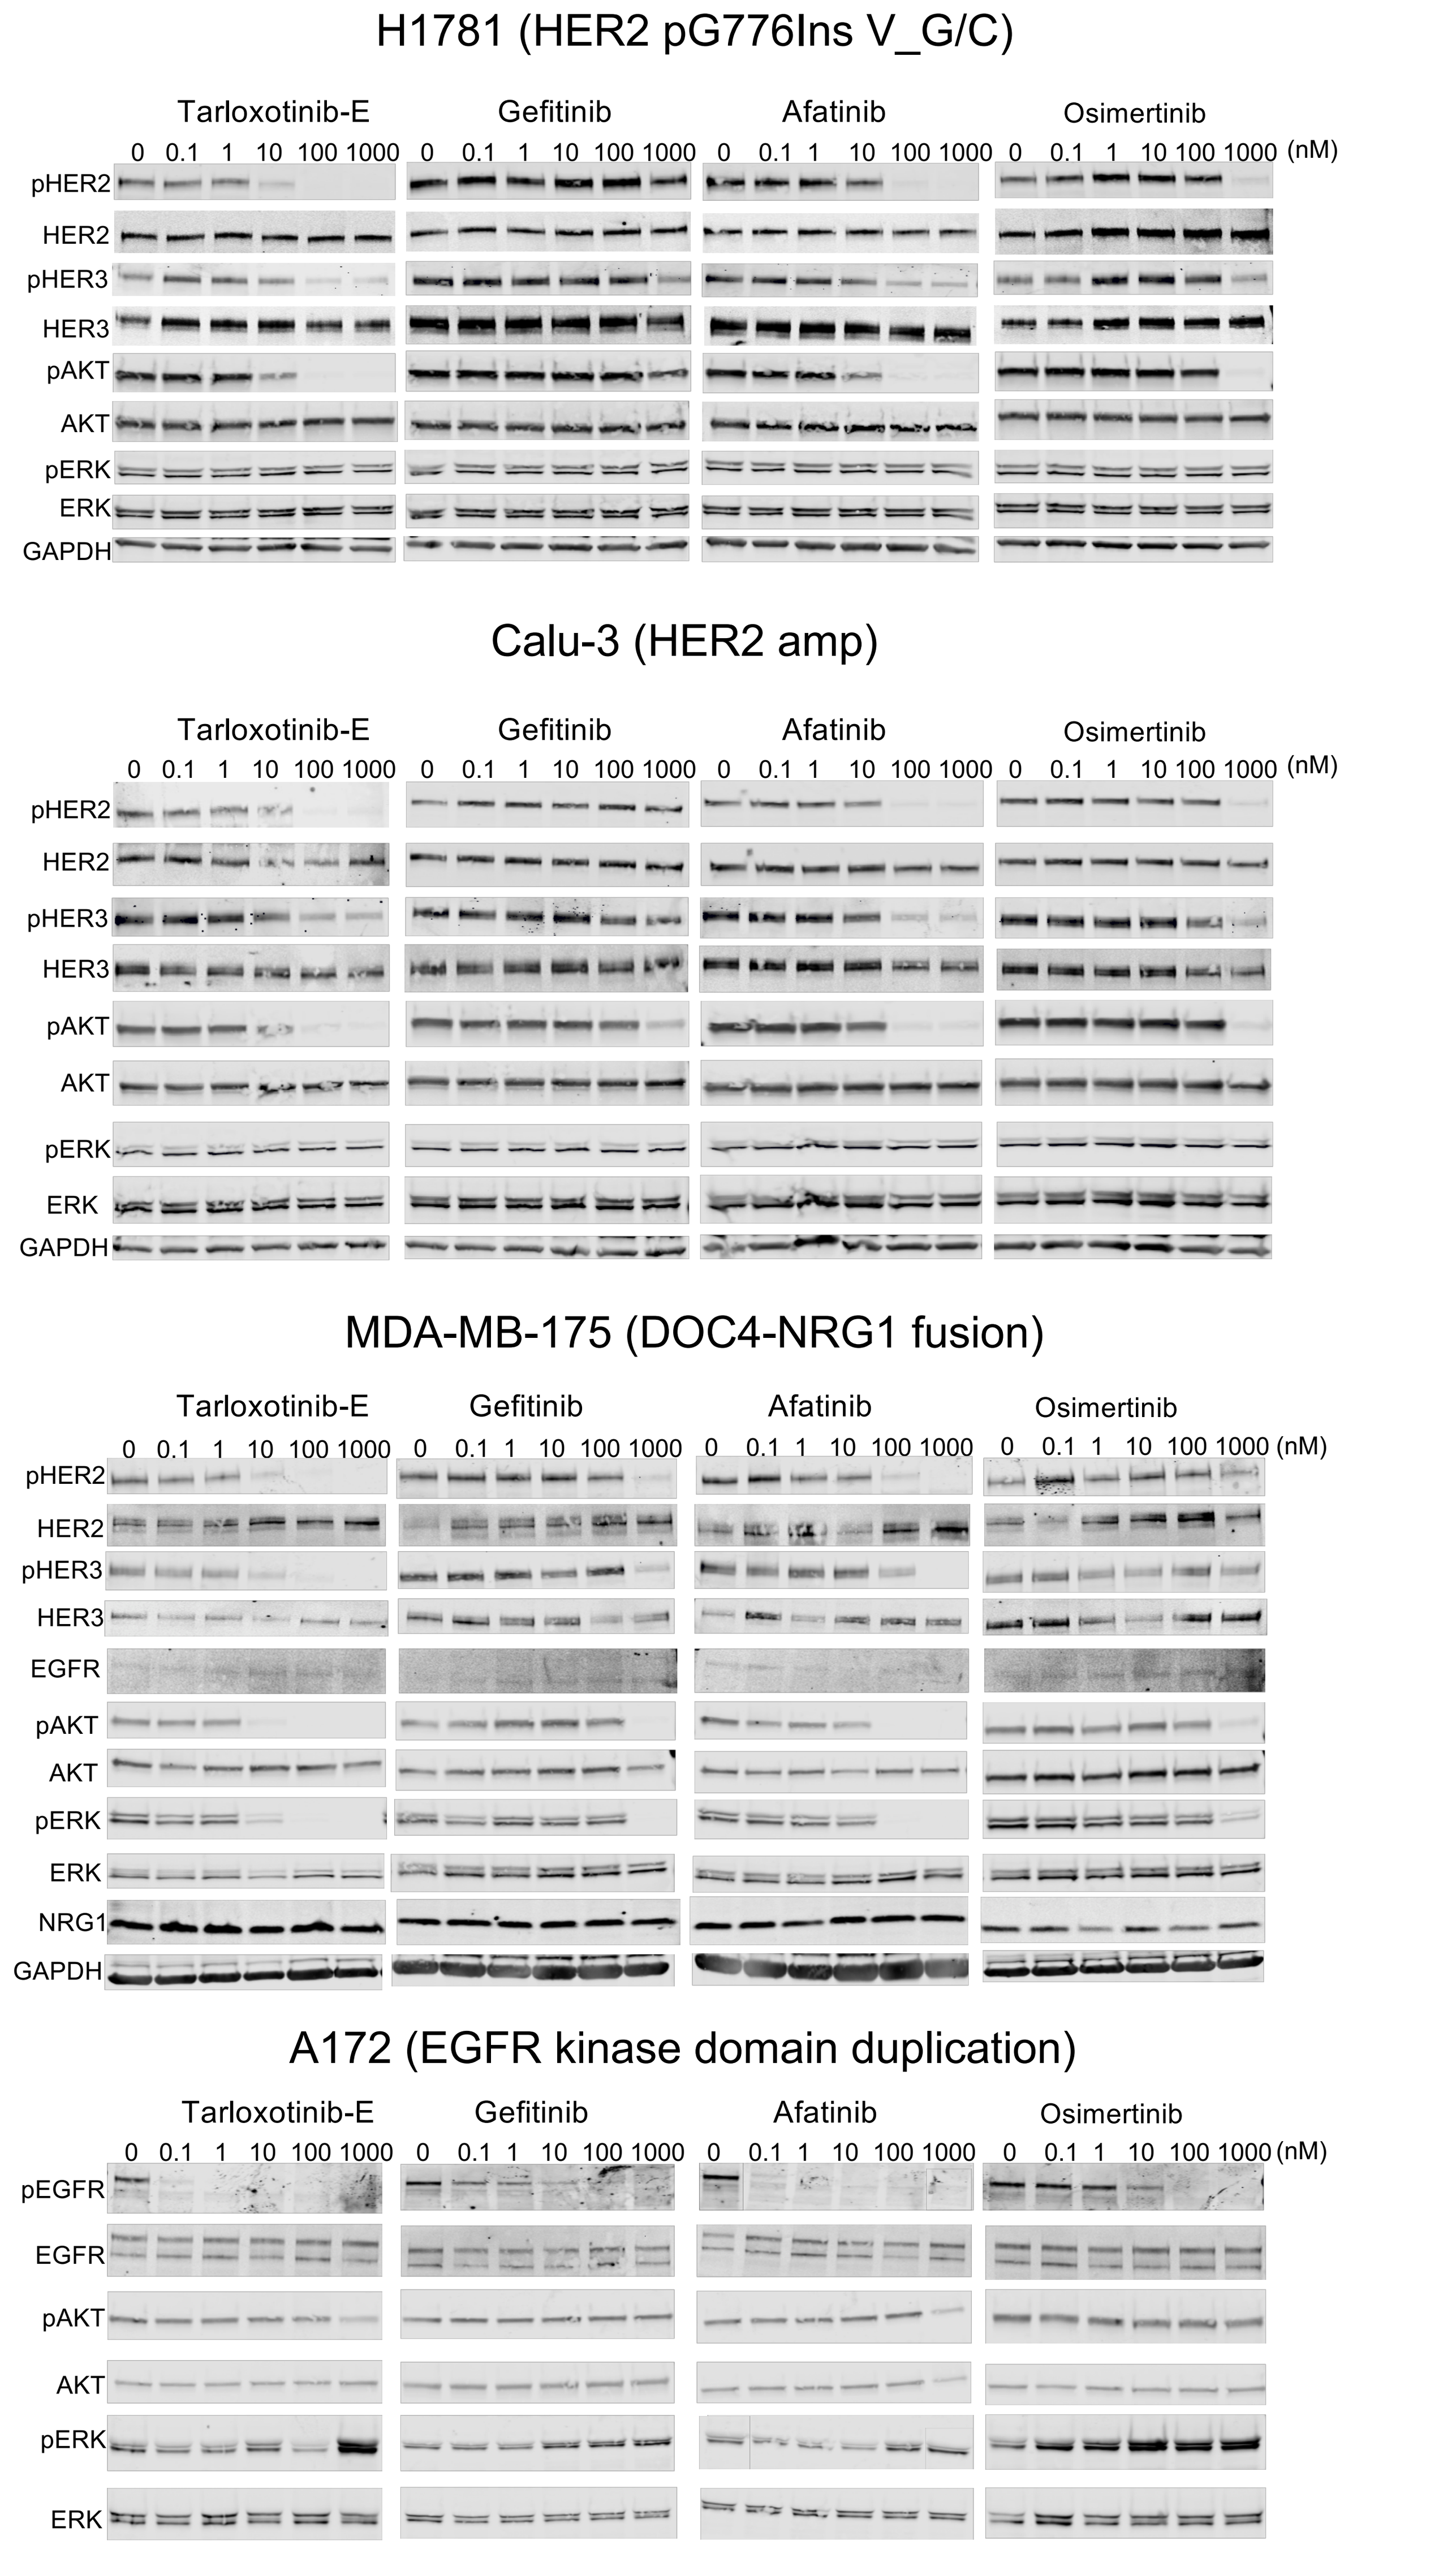
H1781 (HER2 pG776Ins V_G/C)
Calu-3 (HER2 amp)
MDA-MB-175 (DOC4-NRG1 fusion)
A172 (EGFR kinase domain duplication)
Tarloxotinib-E
Gefitinib
Afatinib
Osimertinib
0 0.1 1 10 100 1000 (nM)
pHER2
HER2
pHER3
HER3
EGFR
pEGFR
pAKT
AKT
pERK
ERK
NRG1
GAPDH

Figure 2. Tarloxotinib-E inhibits ErbB family members phosphorylation and signaling in HER2 and EGFR driven cell lines. Cells were treated with the indicated doses of afatinib, gefitinib, osimertinib or tarloxotinib-E (active drug) for 2 hours, lysed and analyzed by immunoblot. Experiments were done in triplicate. Phospho antibodies used: pEGFR (Y1068), pAKT (S473), pERK (Y202/204), pHER2 (Y1221/1222), pHER3 (Y1289).

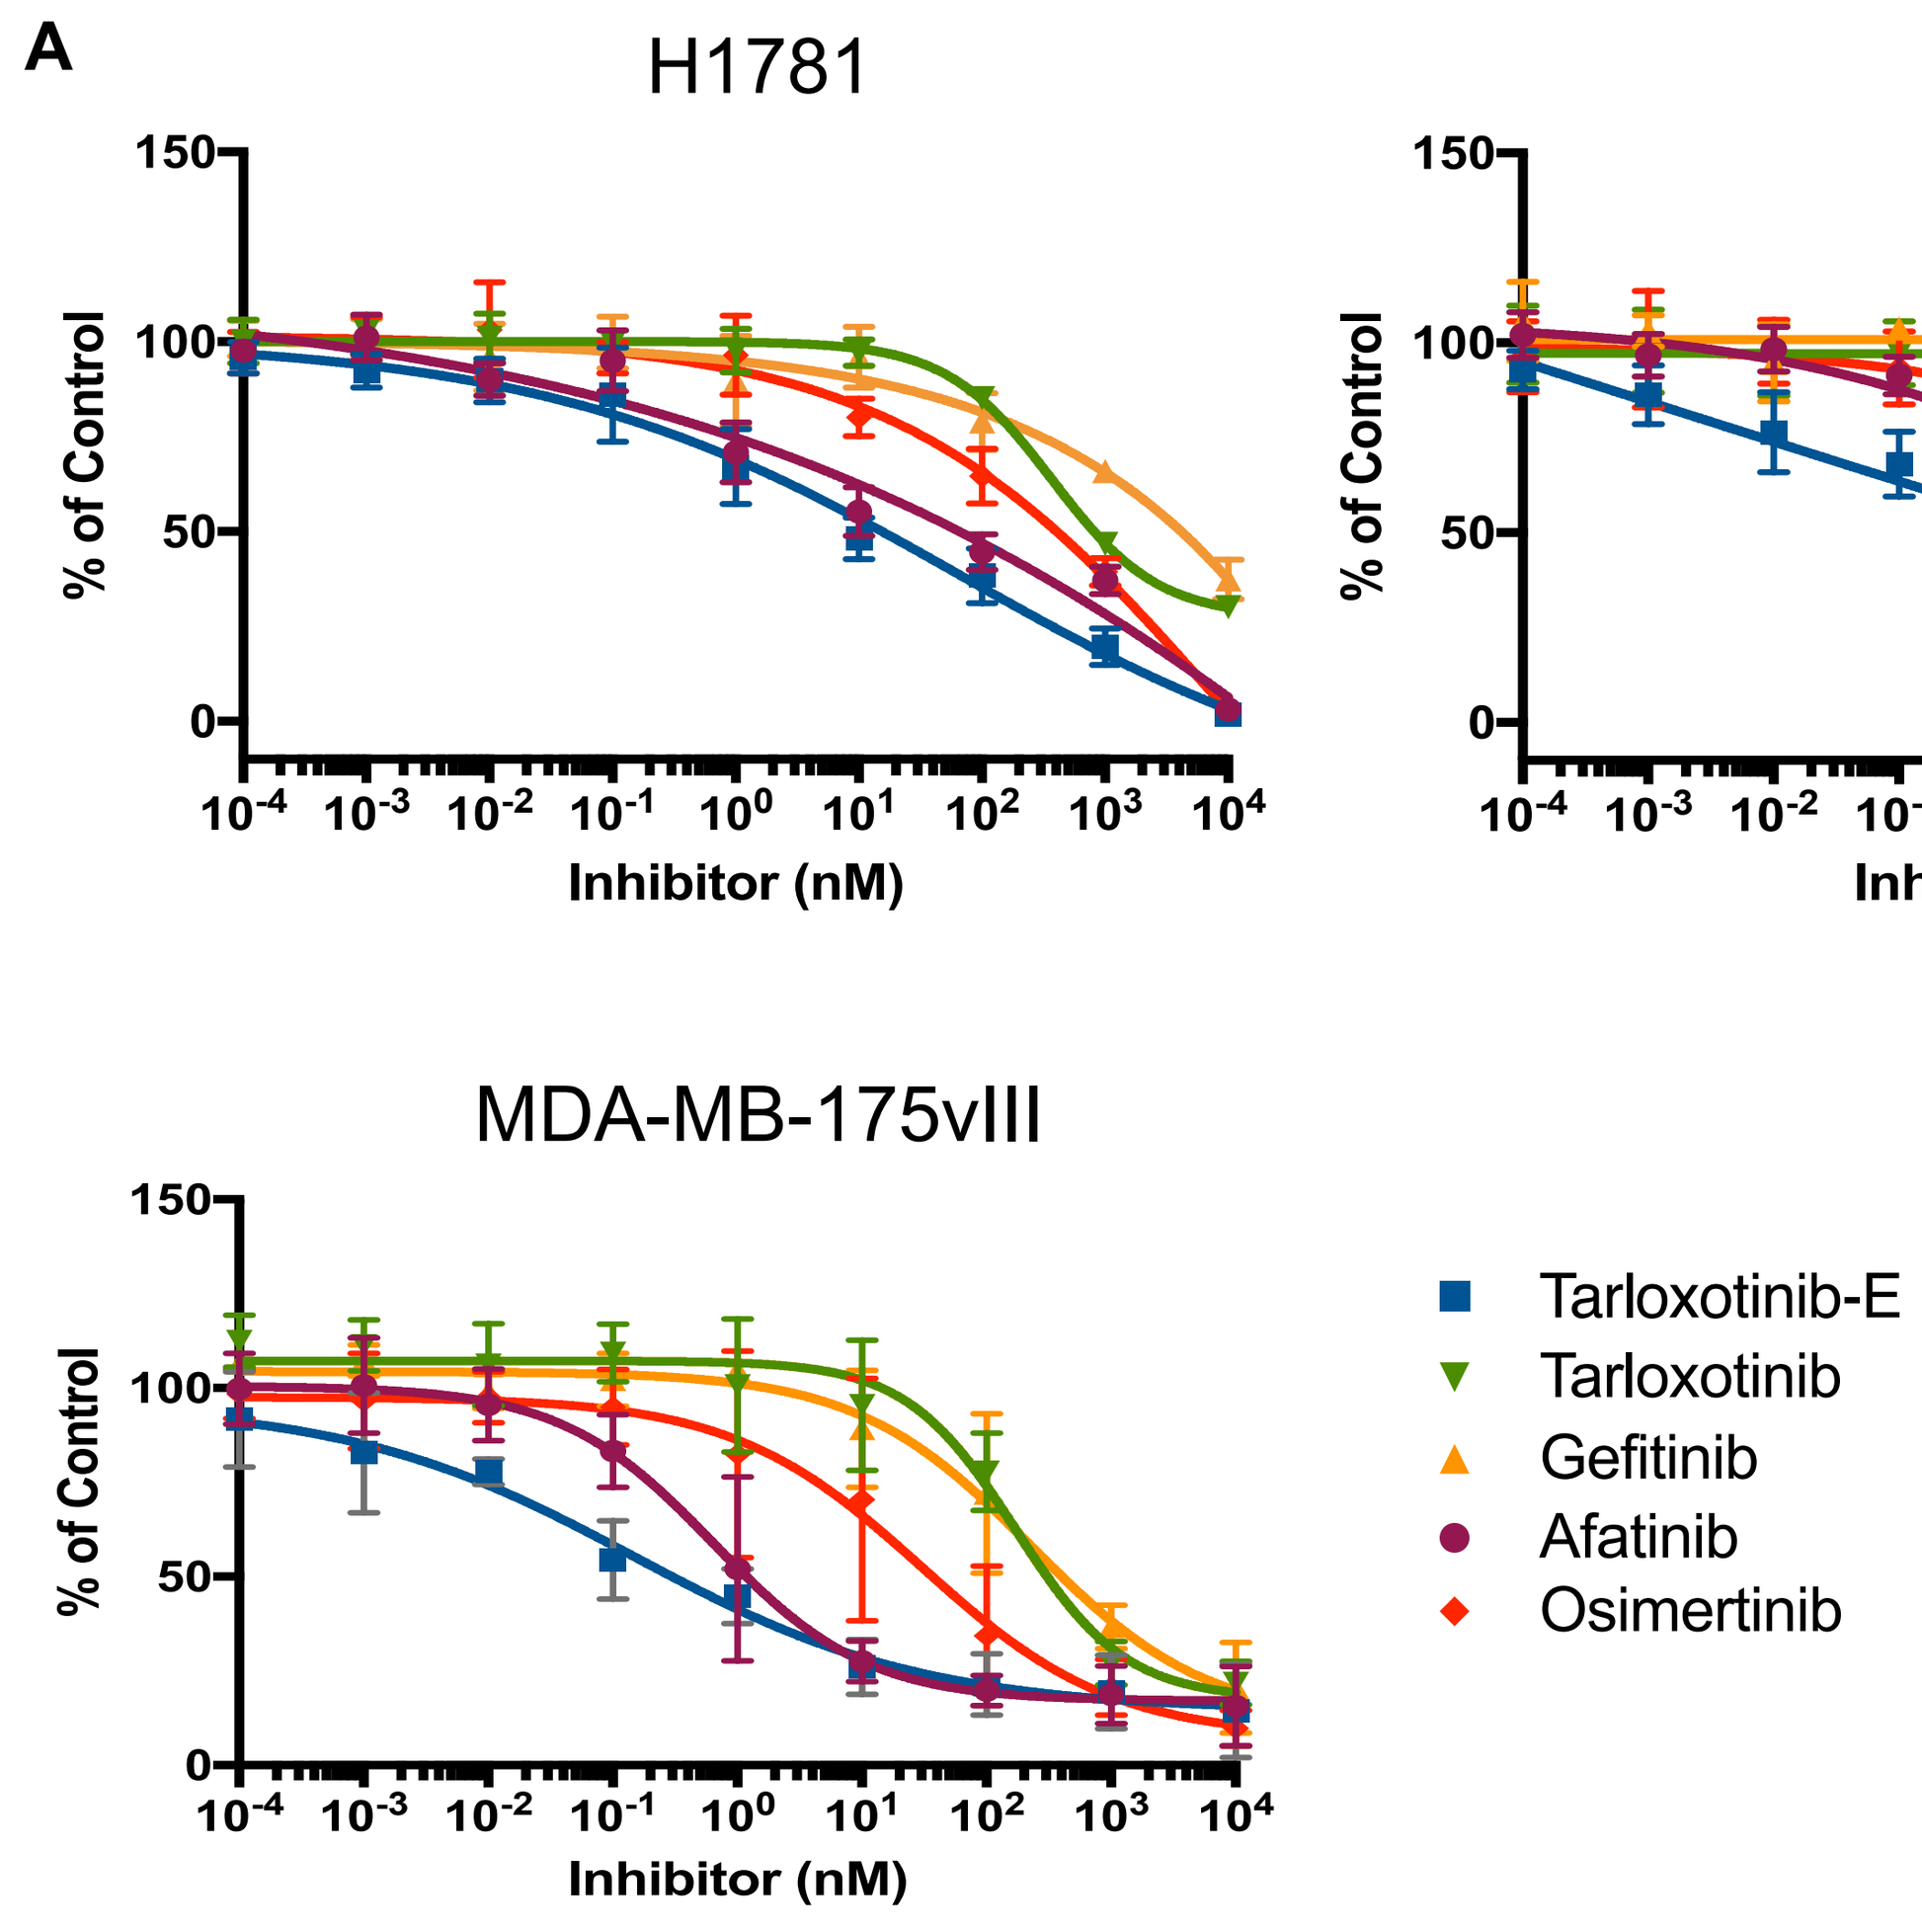
A
H1781
Calu-3
H2170
MDA-MB-175vIII
% of Control
Inhibitor (nM)
Tarloxotinib-E
Tarloxotinib
Gefitinib
Afatinib
Osimertinib

B

	Tarloxotinib-E	Tarloxotinib	Gefitinib	Afatinib	Osimertinib
EGFR-Ex20ins lines					
CUTO-14	72.2	4645	3741	110.9	303
CUTO-17	48.1	3090	4197	219.7	426
CUTO-18	158.4	>10000	>10000	841.3	647
HER2 lines					
H1781 (ex20ins)	15	816	4168	66	406
Calu-3 (amp)	2	325	1324	31	188
H2170 (amp)	4	588	1156	11	113
NRG1 fusion line					
MDA-MB-175vII	0.3	307	404	1.2	37

Figure 3. Tarloxotinib-E inhibits proliferation of cell lines harboring HER2 alterations. A) Dose response curves of cell viability of H1781 (*HER2* ex20ins), Calu-3 (*HER2* amp), MDA-MB-175VIII (Breast, *DOC4-NRG1* fusion) and A172 (Glioma, *EGFR* kinase domain duplication). Cells were treated with afatinib, gefitinib, tarloxotinib (pro-drug) and tarloxotinib-E (active drug) for 72 hours and measured by MTS. Experiments were done in triplicate; mean ± SEM is plotted. **B)** Table summarizing IC_{50} values of the proliferation experiments.

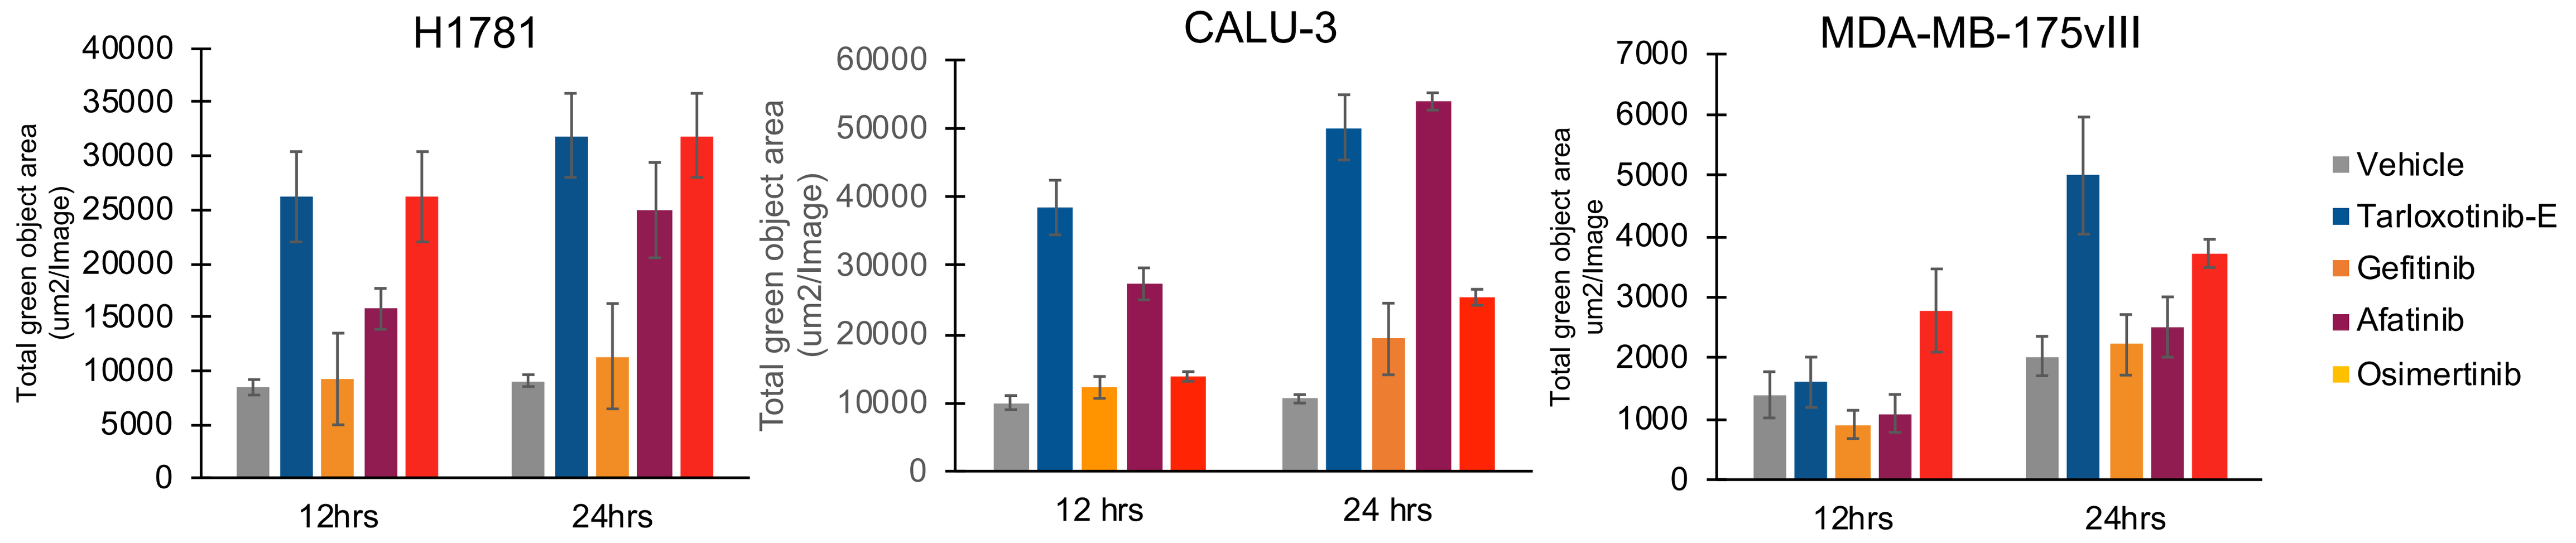
H1781
CALU-3
MDA-MB-175vIII
Total green object area (um2/Image)
12hrs
24hrs
12 hrs
24 hrs
Vehicle
Tarloxotinib-E
Gefitinib
Afatinib
Osimertinib

Figure 4. Tarloxotinib-E induces apoptosis in HER2 altered and NRG1 fusion cell lines. IncuCyte quantification of H1781, CALU-3 and MDA-MB-175vIII cells treated with 10,000nM tarloxotinib-E, gefitinib, afatinib or osimertinib. Apoptotic cells are labeled with caspase-3/7 green reagent and quantified in real time using the IncuCyte live-cell analysis system.

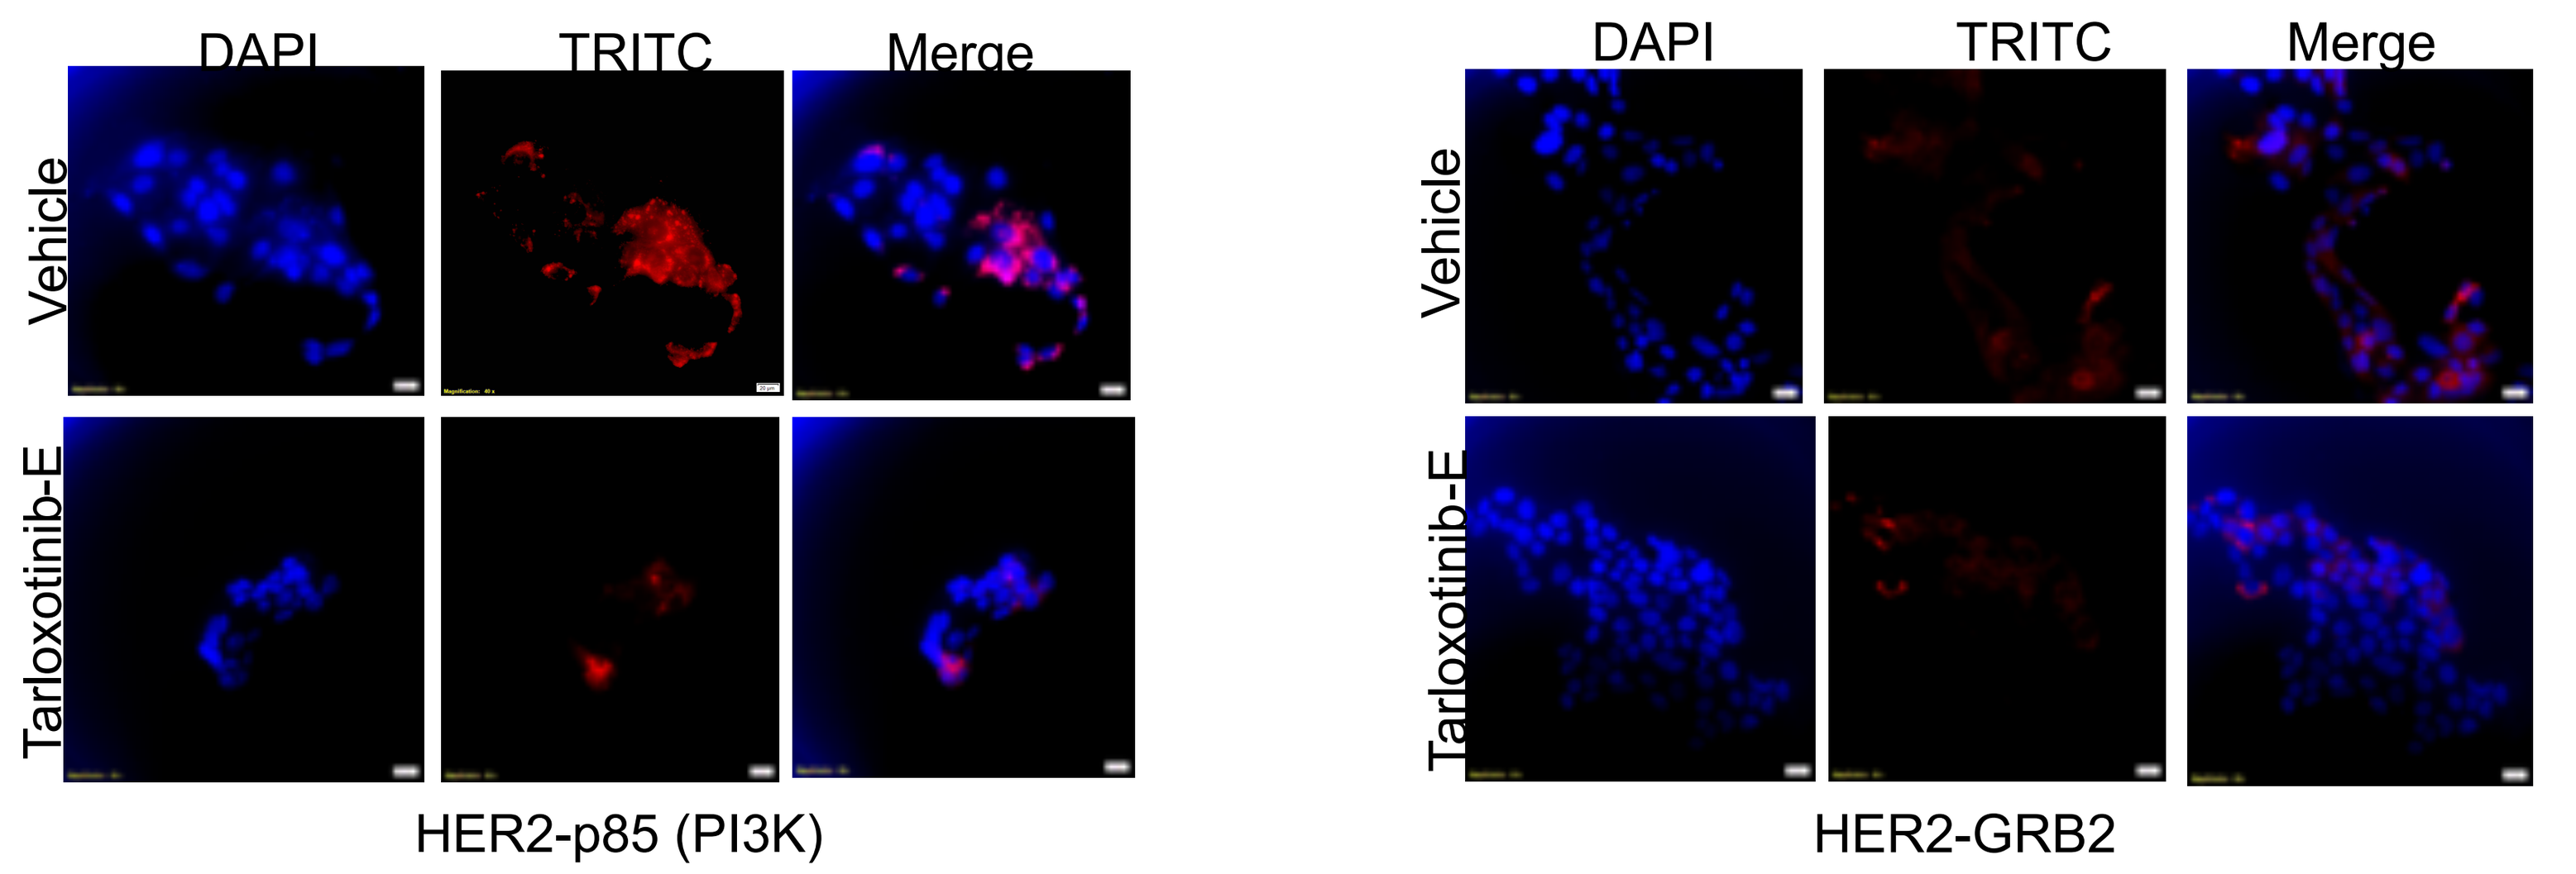
DAPI
TRITC
Merge
Vehicle
Tarloxotinib-E
HER2-p85 (PI3K)
HER2-GRB2

Figure 5. Tarloxotinib-E inhibits interaction of HER2 with subunit p85 of PI3K. Proximity ligation assay was performed in H1781 cells treated with tarloxotonib-E for 30 minutes. Red signal corresponds to positive interaction of HER2 with p85 or GRB2.

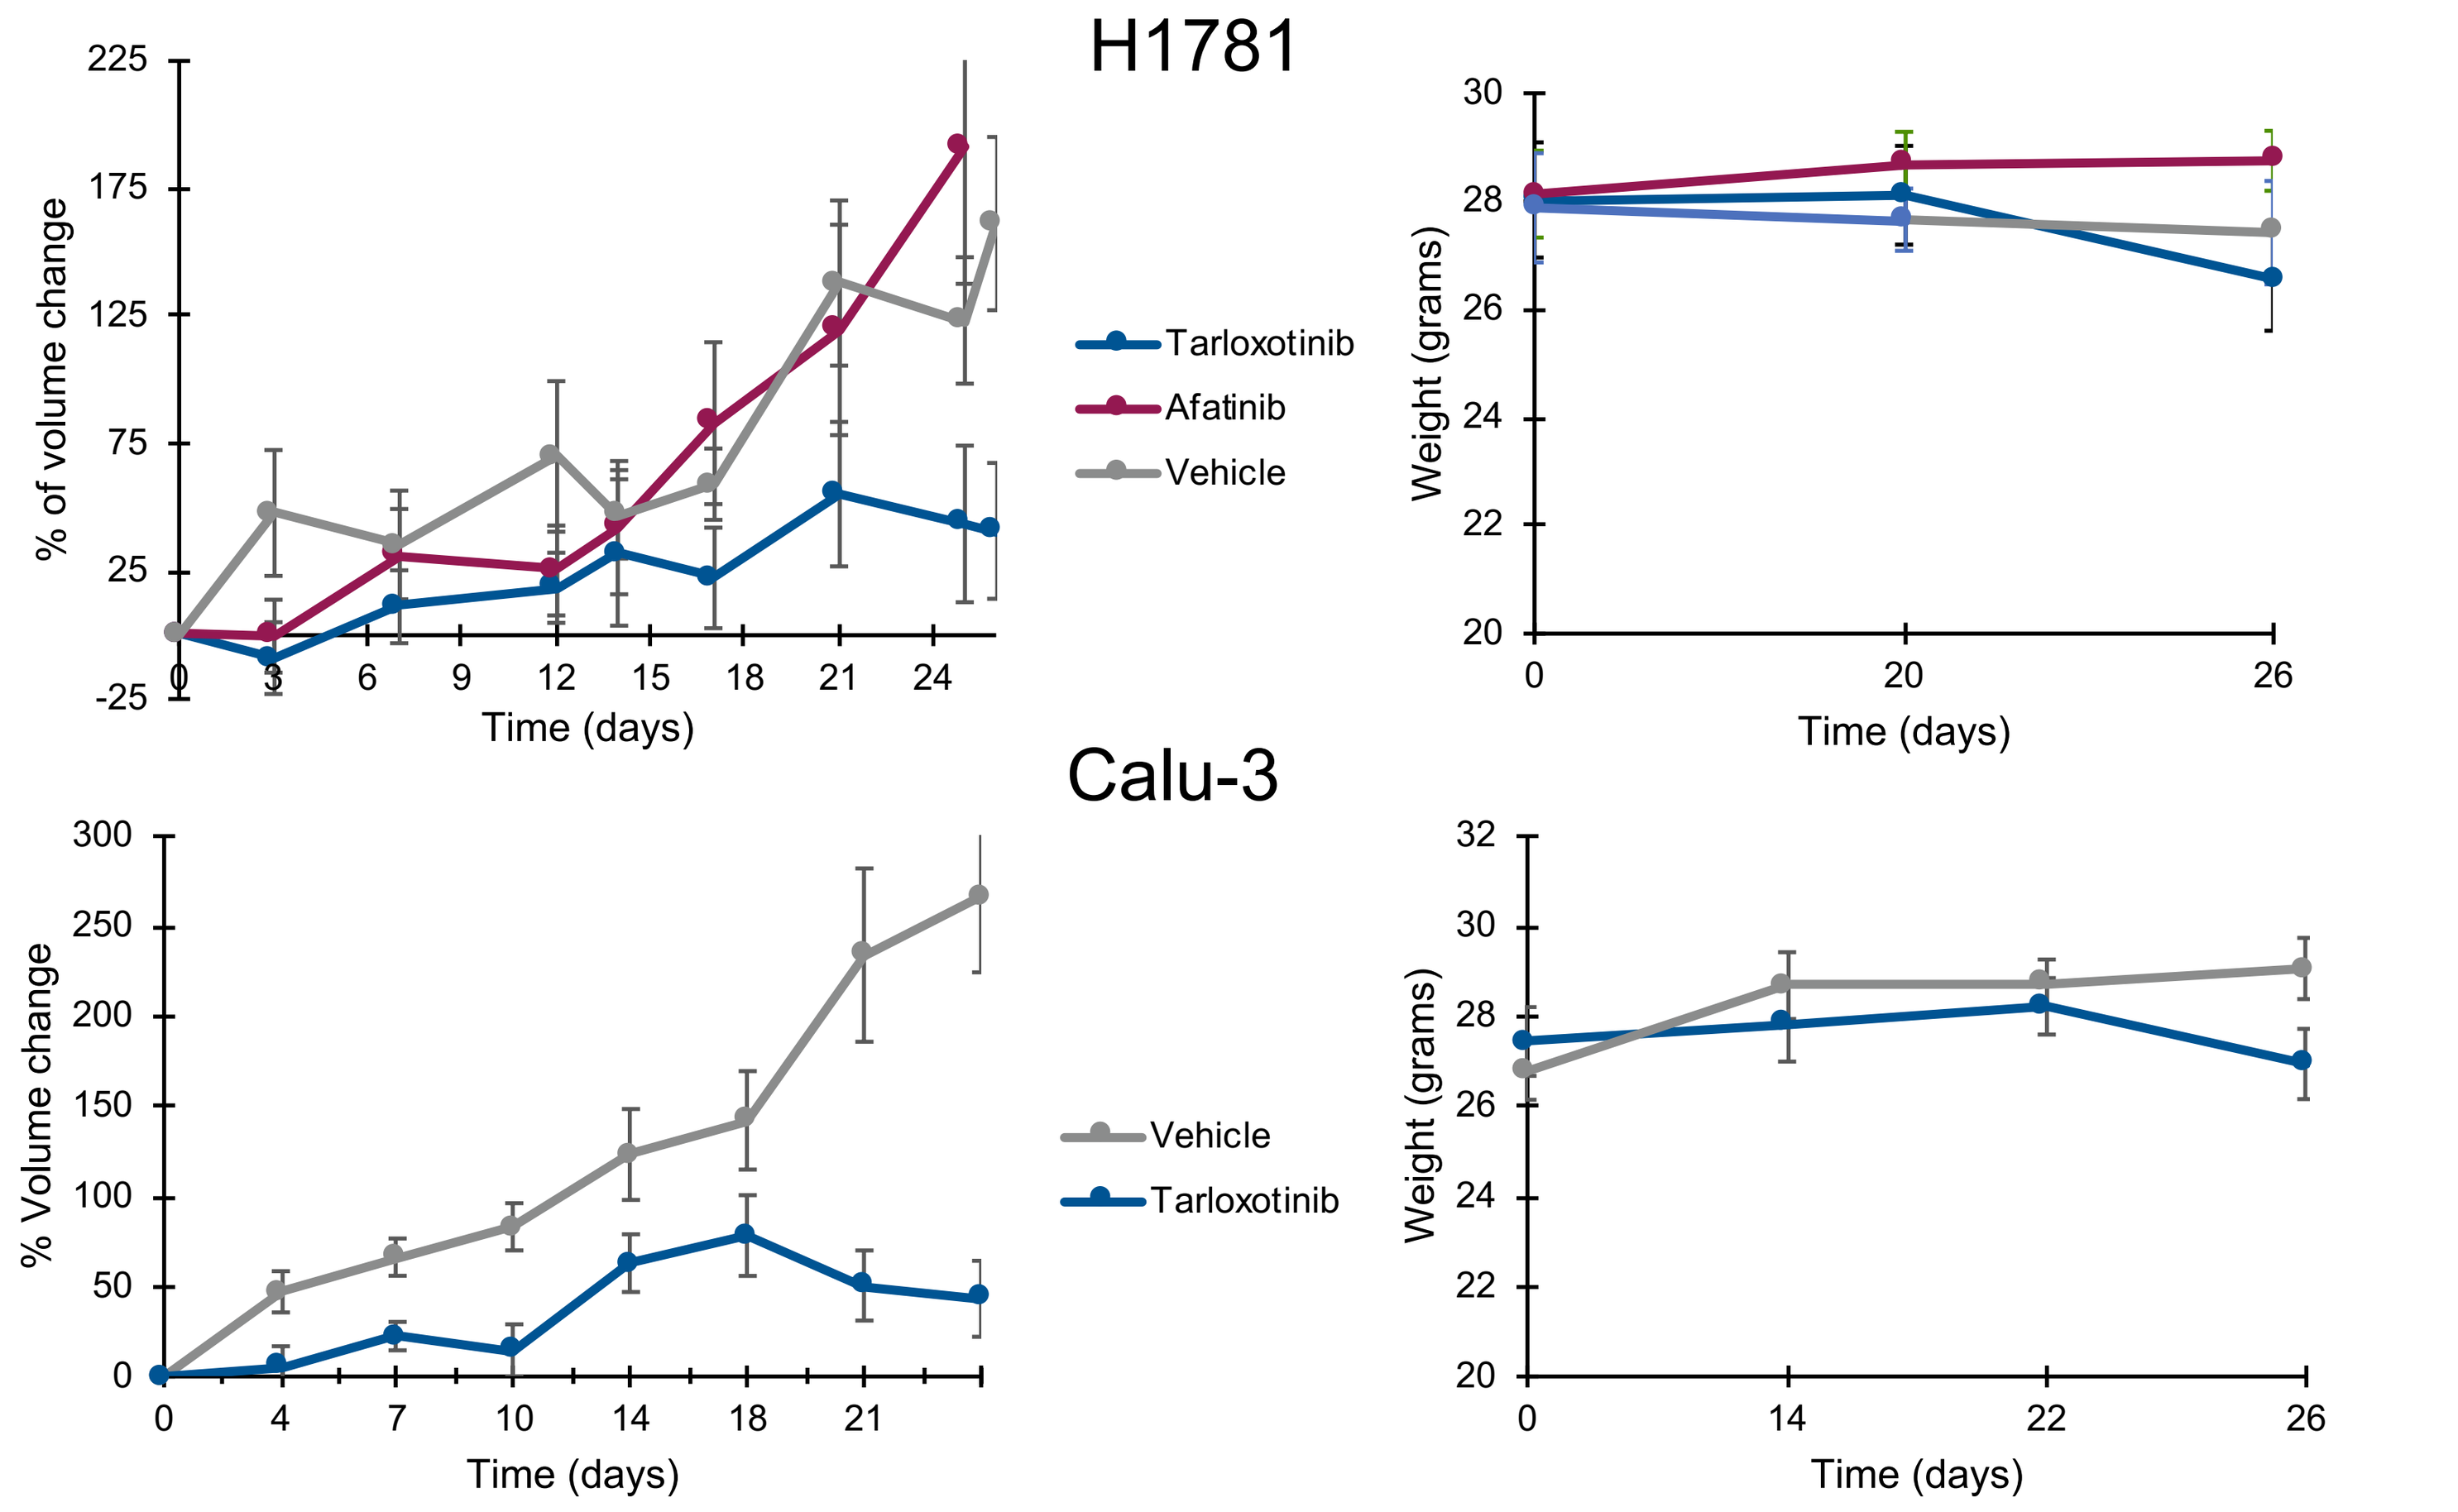
H1781
Calu-3
% of volume change
% Volume change
Time (days)
Weight (grams)
Tarloxotinib
Afatinib
Vehicle

Figure 6. Tarloxotinib inhibits tumor growth of two HER2 dependent xenograft models. Percent changes from baseline tumor volume in nude mice injected subcutaneously with H1781 or Calu-3 cells and treated with vehicle, afatinib (6mg/kg, daily, PO), or tarloxotinib (48mg/kg, once weekly, IP). Graphs on the right show mice weight changes during tarloxotinib treatment.

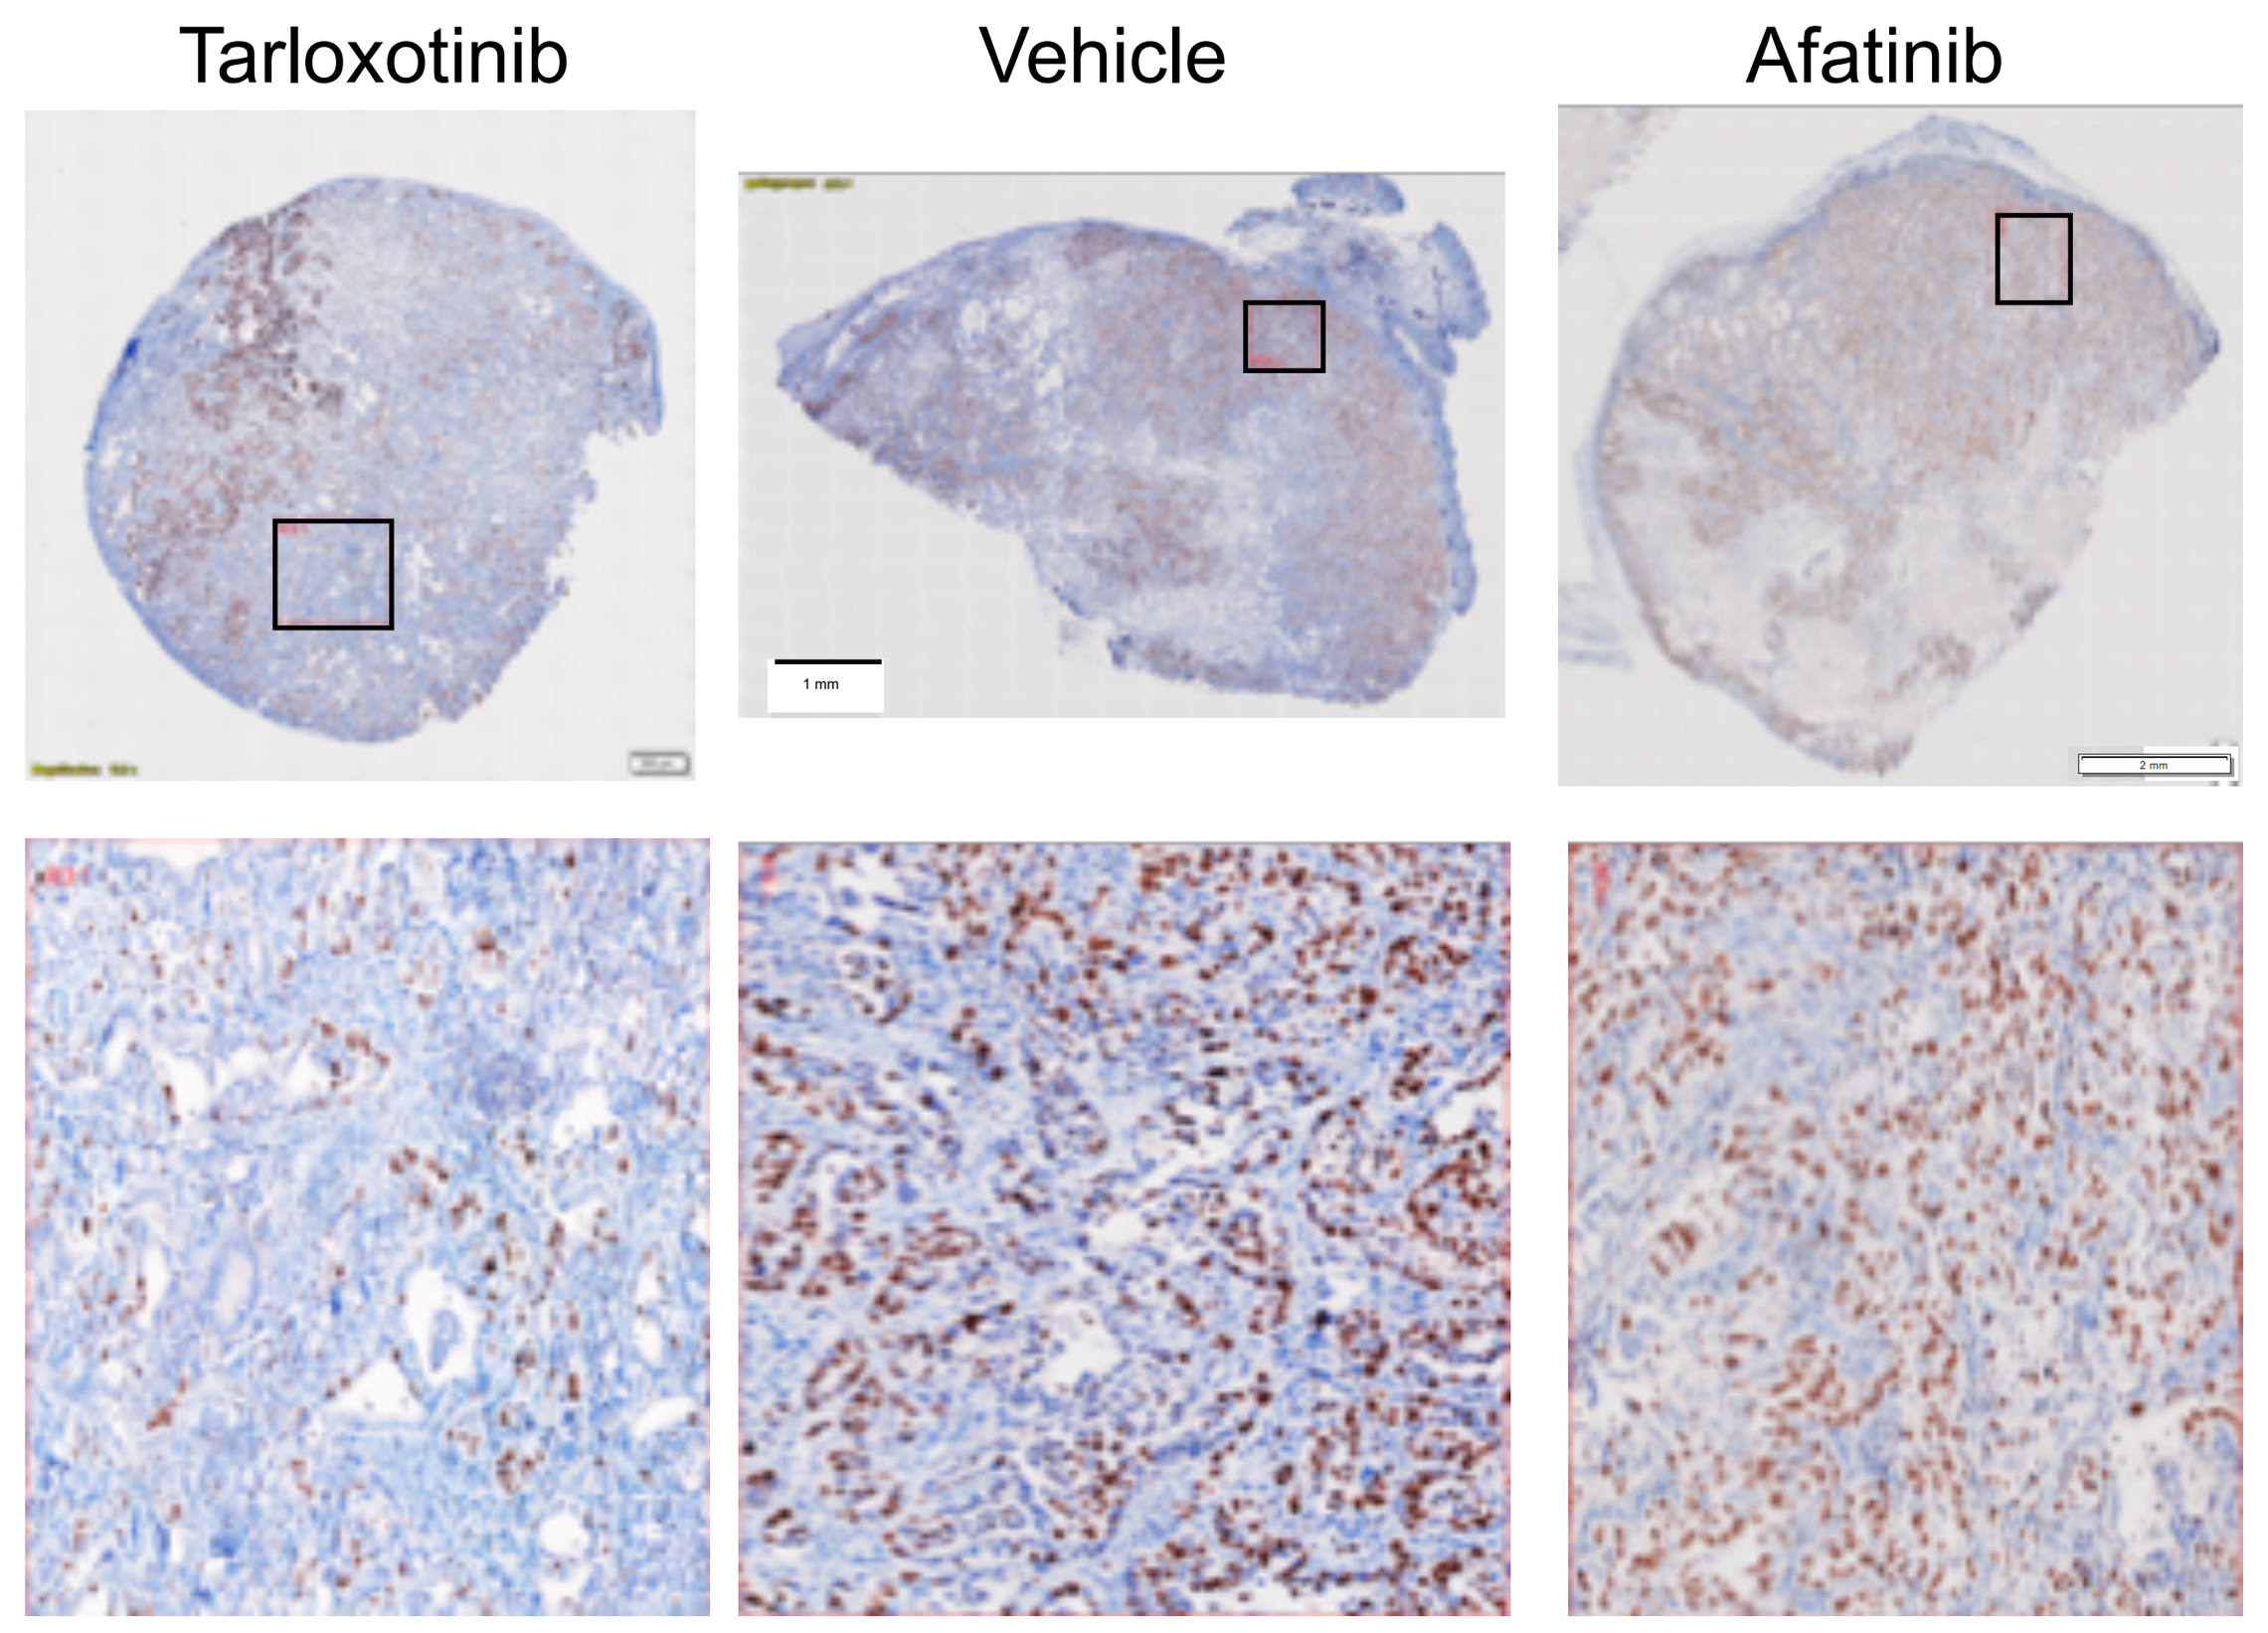
Tarloxotinib
Vehicle
Afatinib

Figure 7. Tarloxotinib inhibits tumor proliferation in H1781 xenograft models. Ki67 IHC was performed in tumors collected from xenografts in figure 6. Bottom panel is a higher magnification of insets.

Conclusions

- Tarloxotinib-E (active drug) inhibits *in vitro* proliferation of HER2 altered cell lines, cells harboring an NRG1 fusion or cells expressing an EGFR kinase domain duplication.
- In HER2 dependent cell lines, tarloxotinib-E has 60-150 fold higher activity compared to tarloxotinib *in vitro* under normoxic conditions.
- Tarloxotinib-E inhibits HER2 and HER3 phosphorylation and downstream signaling *in vitro.* Additionally, tarloxotinib-E inhibits phosphorylation of EGFR in an EGFR kinase duplication cell line.
- HER2 altered cell lines favor PI3K/AKT signaling axis over MAPK signaling.
- The active drug induces apoptosis after 12-24 hours of exposure.
- Tarloxotinib significantly reduces tumor volume and proliferation in two HER2 xenografts models.

References

1. Siegel RL, et al. CA Cancer J Clin. 2016;66:7–30. 2. Pillai RN, et al. Cancer. 2017;123:4099-4105. 3. Arcila ME, et al. Clin Cancer Res. 2012;18:4910-4918. 4. Gay ND, et al. J Thorac Oncol. 2017;12:E107-E110. 5. Gallant JN, et al. Cancer Discov. 2015;5:1155-1163. 6. Herter-Sprie GS, et al. Front Oncol. 2013;3:86. 7. Zegers CML, et al. Radiother Oncol. 2013;109:100-106. 8. Silva S, et al; Poster presented at AACR-NCI-EORTC International Conference. Molecular Targets and cancer therapeutics. November 5-9, 2015. Boston, MA.

Funding: Developmental Therapeutics Program, University of Colorado Cancer Center (NIH/NCI P30CA046934).